ACCELERATING BONE HEALING USING A NOVEL SCLEROSTIN INHIBITOR

H. Rosenthal[1] , F.S. Kimball[2], S.H. Berven[3], H.M. Aberman[4], M.J. Larson[5], D.L. Ellies[2]
[1] Medical Director Sarcoma Division, Associate Professor of Orthopaedic Surgery, University of Kansas Health System, KS.
[2] Ossifi Inc, Overland Park, KS.
[3] Department of Orthopeadic Surgery, UCSF, CA.
[4] Safety and Efficacy Advisors, Sewalls Point, FL
[5] Ibex Preclinical Research, Inc., Logan, UT.

Despite the successful development of various bone void fillers, they all appear to have similar properties, which do not accelerate the bone healing process unless they carry an osteoinductive component, like InFuse™. Very few bone void fillers have an osteoinductive component, and all of the osteoinductive agents are biologic in nature. Here we present a novel small molecule capable of significantly accelerating osteoinduction to result in 2x increase in new mature bone in 5 weeks, which has been shown, in vitro, to be equal to using 10 units of InFuse in a single dose.

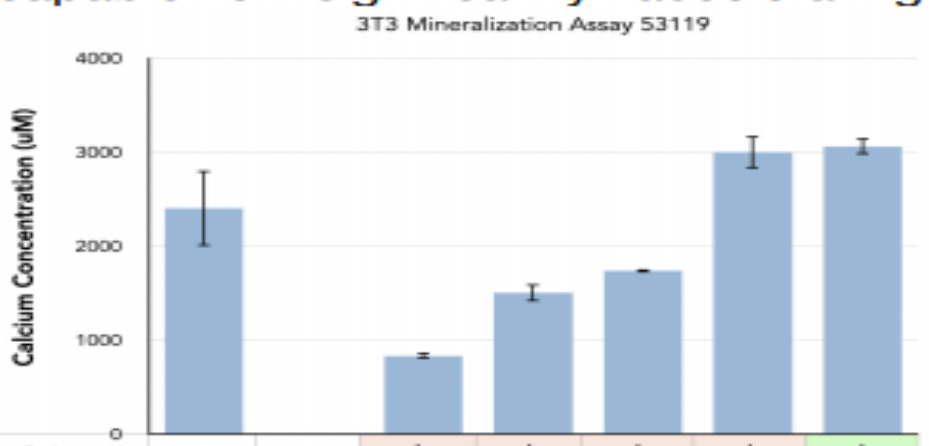
3T3 Mineralization Assay 53119
Calcium Concentration (uM)
4000
3000
2000
1000
0

Sclerostin		+	+	+	+	+
Optimized O1-1						+
InFuse™ Col1		+				+
InFuse™ BMP			0.1X	1X	10X	

Sclerostin, the product of the SOST gene, is secreted by osteocytes and antagonizes the Wnt/LRP pathway, which is essential for osteoblast proliferation, differentiation and mineralization. Humans devoid of SOST present with 5x increase in bone density and no other clinical pathology (Brunkow et al. Am. J. Hum. Gen. 2001). Mice that have been engineered to overproduce Sclerostin protein result in animals displaying osteoporotic bones (Li et al, JBMR 2008). Sclerostin, in its natural state, is a negative regulator of bone formation, so by inhibiting the inhibitor, one increases bone formation. The mammalian skeleton consists of bones that are formed primarily by two different ways; long bones via endochondral ossification, whereas flat bones are formed via intramembranous ossification. The major difference is that in endochondral ossification, the bones are formed using a cartilage intermediary similar to that found when stimulating the BMP pathway.

The objective of this study was to determine the efficacy of a sclerostin small molecule inhibitor, OssiFi (OFX) O1-1, combined with a synthetic bone graft (MG), in a rabbit intramembranous calvarial ossification model. In an effort to provide a source of cells to respond to the sclerostin inhibitor, we created four 7mm calvarial defects in each rabbit calvaria that did not penetrate the basal plate (Kim et al., J. Biomed Mater. Res. 2019). Each animal received one synthetic bone graft alone (negative control), and three synthetic bone grafts with OFX O1-1 at different doses. The animals did not present with any abnormalities during the course of the study (5 weeks). At 3 weeks we found that the synthetic bone void filler (MG) alone left a sizable defect, ~60%, but the OFX O1-1/MG combination reduced the defect size dose dependently to 13%. At 5 weeks the MG synthetic bone void filler alone resulted in 58% new bone in the filled defect, whereas the mean result of OssiFi's OFX O1-1/MG, dose dependently, filled the defect to 95% with new bone.

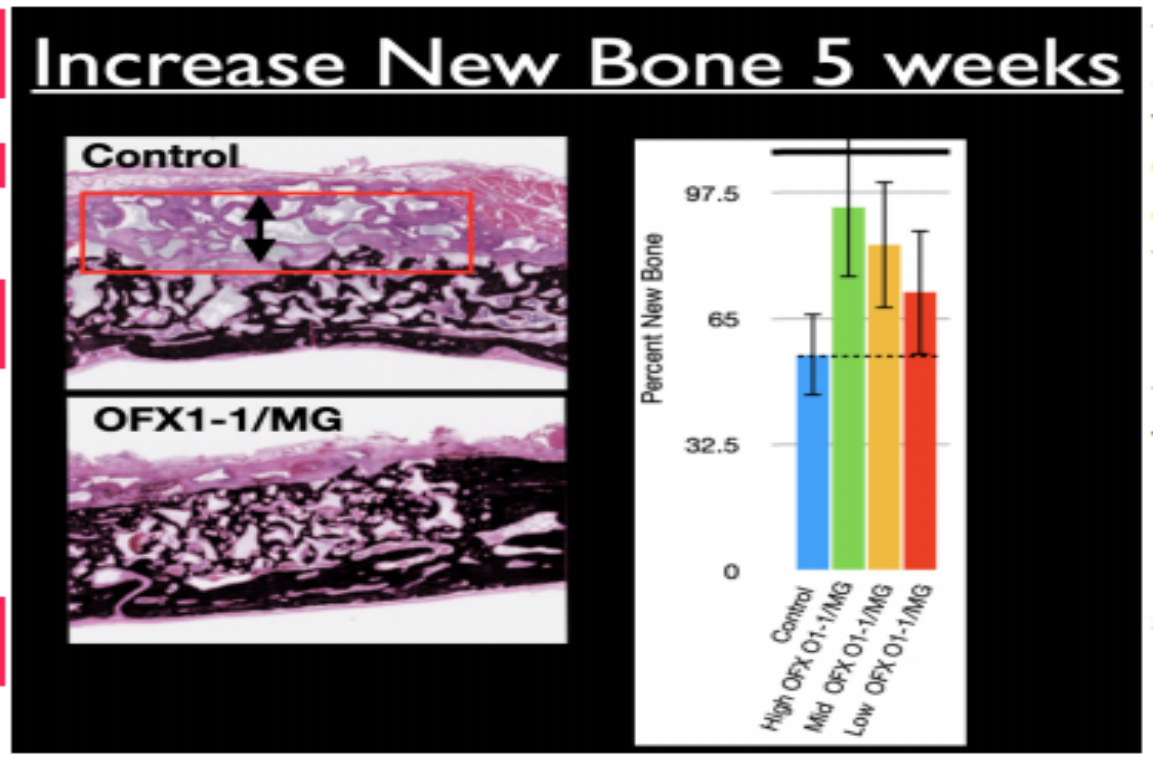
Increase New Bone 5 weeks
Control
OFX1-1/MG
Percent New Bone
97.5
65
32.5
0
Control
High OFX O1-1/MG
Mid OFX O1-1/MG
Low OFX O1-1/MG

In summary, we found at 5 weeks, in a rabbit 7mm calvarial defect, that the defect was not visible to the naked eye displaying good osteo-integration, mature woven vascularized bone, and 2x more new bone than the synthetic bone void filler MG, alone.